A Phase I/II, Dose-Escalation Study of Daratumumab, a CD38 mAb in Patients with Multiple Myeloma – Preliminary Safety Data

Peter Gimsing[1], Torben Plesner[2], Hareth Nahi[3], Henk Lokhorst[4], Marie-Louise Valentin[5], Steen Lisby[5], Paul Richardson[6]

[1]Hematology, Copenhagen University Hospital, Copenhagen, Denmark; [2]Hematology, Vejle Hospital, Vejle, Denmark; [3]Internal medicine / Hematology, Karolinska Institutet, Stockholm, Sweden; [4]Hematology, University Medical Center Utrecht, Utrecht, Netherlands; [5]Genmab A/S, Copenhagen, Denmark; [6]Medical Oncology, Dana-Farber Cancer Institute, Boston, MA, USA.

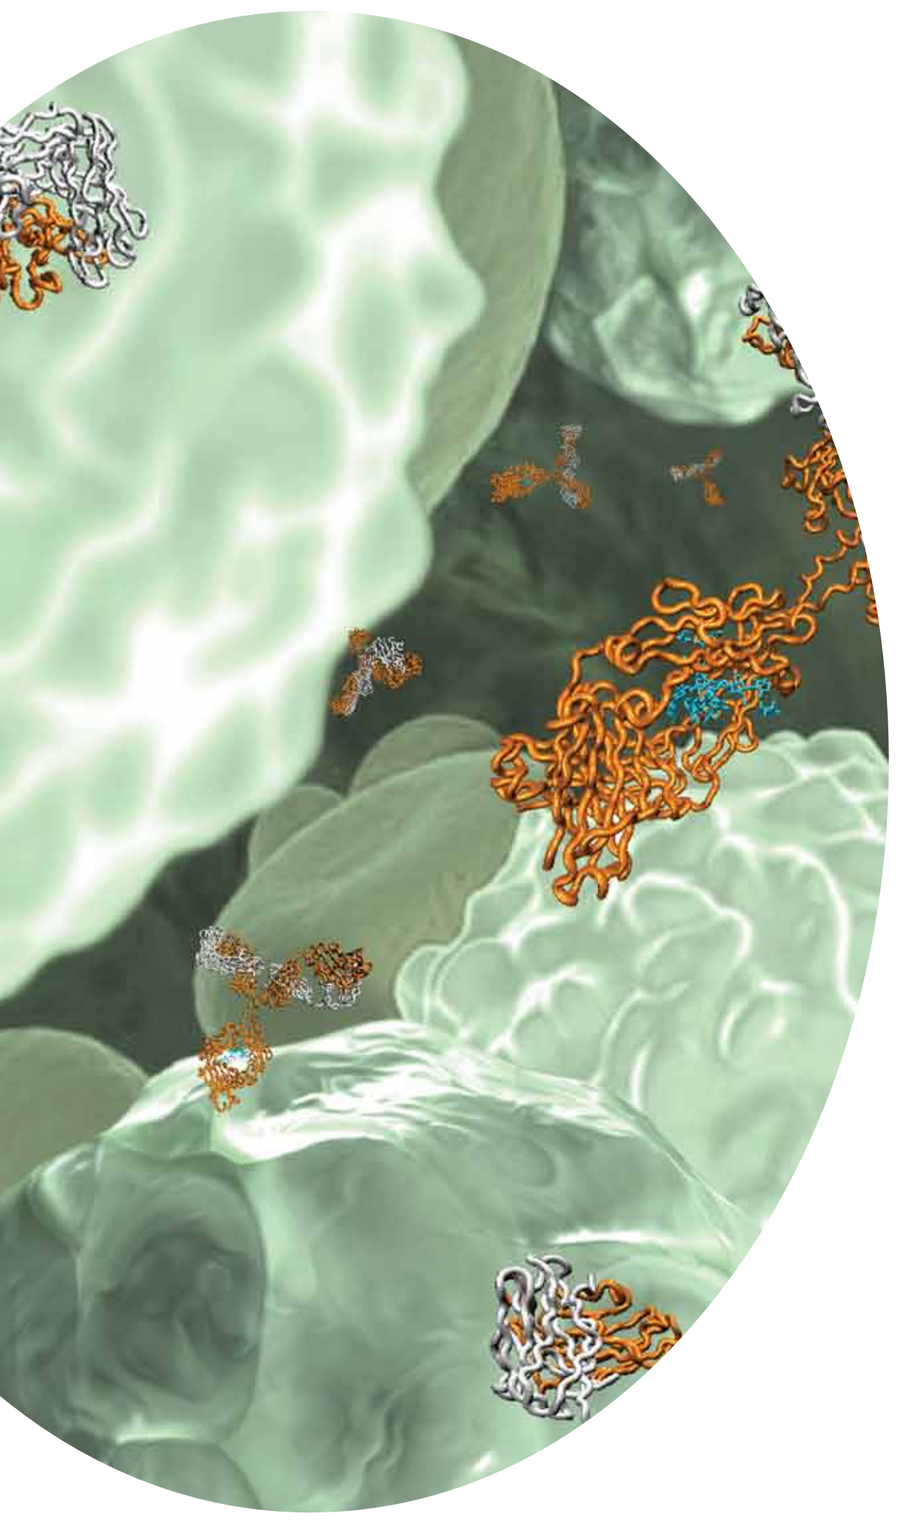

BACKGROUND

Multiple myeloma (MM) is an uncontrolled and progressive proliferation of a plasma cell clone. The proliferating MM cells displace the normal bone marrow leading to dysfunction in normal hematopoietic tissue and destruction of the normal bone marrow architecture. It accounts for approximately 1% of all malignancies and 10% of all hematological malignancies. Currently, no cure is available for patients with this malignant disease and those who relapse or are refractory to the current treatment options have a poor prognosis. Therefore, new treatment options are highly needed for this patient population.
MM cells exhibit a very strong expression of CD38 transmembrane glycoprotein molecules suggesting this could be an optimal therapeutic target for antibody therapy. Daratumumab is a human CD38 monoclonal antibody with broad-spectrum killing activity. Daratumumab mediates MM cell death via antibody-dependent cellular cytotoxicity and complement-dependent cytotoxicity[1], which are the major mechanisms of action. Daratumumab has been shown to inhibit growth of CD38-expressing tumor cells in SCID mouse xenografts[1].
We report the preliminary safety and efficacy results from the ongoing first-in-human dose-escalation study (ClinicalTrials.gov NCT00574288).

OBJECTIVES

Primary objective:

- Establishment of the safety profile of daratumumab when given as monotherapy in patients with MM relapsed or refractory to at least two different prior therapies.

Secondary objectives include:

- Establishment of maximal tolerated dose (MTD) for future studies with daratumumab.
- Evaluation of efficacy of daratumumab when given as monotherapy in patients with MM.

METHODS

This first-in-human study consists of two parts: Part 1 dose-escalation to define the MTD; Part 2 cohort expansion of MTD or maximal dose from Part 1 using an extended dose-schedule for 24 weeks. Eligible patients are ≥18 years, diagnosed with MM, ineligible for ASCT, requiring systemic therapy and considered relapsed or refractory to at least two different prior therapies. Patients receive 2 pre- and 7 full-dose infusions of daratumumab over a 9 weeks period: 1st pre-dose day 1, 1st full-dose day 2, 2nd pre-dose day 21, 2nd full-dose day 22 and thereafter weekly full-dose daratumumab. The study design encompasses an accelerated dose-escalation: 3 + 3 patients per cohort with the possibility to limit exposure to 1 patient in the first 2 cohorts depending on safety data. The doses range from 0.005 mg/kg to 24 mg/kg. Pre-medication consisting of steroids, acetaminophen and antihistamine is administrated before all infusions.
AEs are rated according to CTCAE[2] version 3.0. A Critical Adverse Event (CAE) is defined as: adverse drug reactions (ADRs) preventing the infusion from being resumed and serious infusion-related ADRs ≥ grade 2 (not resolving within 6 hours), non-infusion-related ADRs ≥ grade 3, hemolysis ≥ grade 3 or serious ≥ grade 2, and other serious hematological ADRs ≥ grade 2. When an AE meets the CAE criteria, the Independent Data Monitoring Committee (IDMC) reviews data and determines if the CAE constitutes a Dose Limiting Toxicity (DLT). SAEs and non-serious AEs grade 3 are reported within 24 hours to Genmab A/S.
Cut-off date for SAEs, non-serious grade 3 AEs and M-component is 24-Nov-2011 and cut-off date for other AEs is 18-Oct-2011. The results presented are based on preliminary data before database lock.

RESULTS

Safety data for 23 patients, including dose range from 0.005 mg/kg up to and including 4 mg/kg cohort, were collected. Five SAEs were assessed as related to daratumumab. One patient had anemia grade 3 and thrombocytopenia grade 4, one patient had increased AST grade 3 and based on these DLT events 3 more patients were enrolled in the 0.1 mg/kg and 1.0 mg/kg cohorts. One patient experienced a grade 3 bronchospasm and one patient experienced cytokine release syndrome grade 2. All patients recovered after relevant treatment.
Since implementation of pre-medication before all infusions, steroids 2 days following infusion and dilution of trial drug, no serious infusion-related AEs have been reported.

Characteristic	<=0.5 mg/kg (N=11)	1 mg/kg (N=6)	2 mg/kg (N=3)	4 mg/kg (N=3)	Total (N=23)
Gender, n					
Male/Female	7/4	3/3	3/0	1/2	14/9
Age, years					
Median (range)	62 (42-76)	63 (45-71)	64 (60-71)	64 (62-66)	64 (42-76)
Prior therapeutic regimens					
Median (range)	6 (2-9)	5.5 (3-8)	8 (6-10)	6 (3-6)	6 (2-10)
Years since MM diagnosis					
Median (range)	5.0 (1.8-12.3)	5.1 (2.7-9.1)	8.4 (6.6-8.9)	5.7 (4.7-12)	5.7 (1.8-12.3)
Body weight					
Median (range)	78 (60-90)	85 (67-117)	94 (92-108)	70 (68-71)	80 (60-117)
ECOG PS					
0	5	0	0	2	7
1	6	5	3	1	15
2	0	1	0	0	1

Table 1: Baseline characteristics and demography.

(Number of events)	<=0.5 mg/kg (N=11)	1 mg/kg (N=6)	2 mg/kg (N=3)	4 mg/kg (N=3)	Total (N=23)
Patients	11	6	3	3	23
Related non-serious grade 3 AEs	3	0	0	2	5
Related SAEs	3	1	1	0	5

Table 2: Overall summary of related AEs.

(% of patients)	<=0.5 mg/kg (N=11)	1 mg/kg (N=6)	2 mg/kg (N=3)	4 mg/kg (N=3)	Total (N=23)
Pyrexia	55	17	0	0	30
Cough	36	0	33	0	22
Free hemoglobin	45	0	0	0	22
Anemia	36	17	0	0	22
Dizziness	18	17	0	33	17
Hemolysis	18	33	0	0	17
Flu-like illness	0	33	33	0	13
Nausea	18	0	0	33	13
Lymphopenia	0	33	33	0	13
Monocytopenia	0	33	33	0	13

Table 3: Incidence of most frequent AEs reported in > 2 patients.

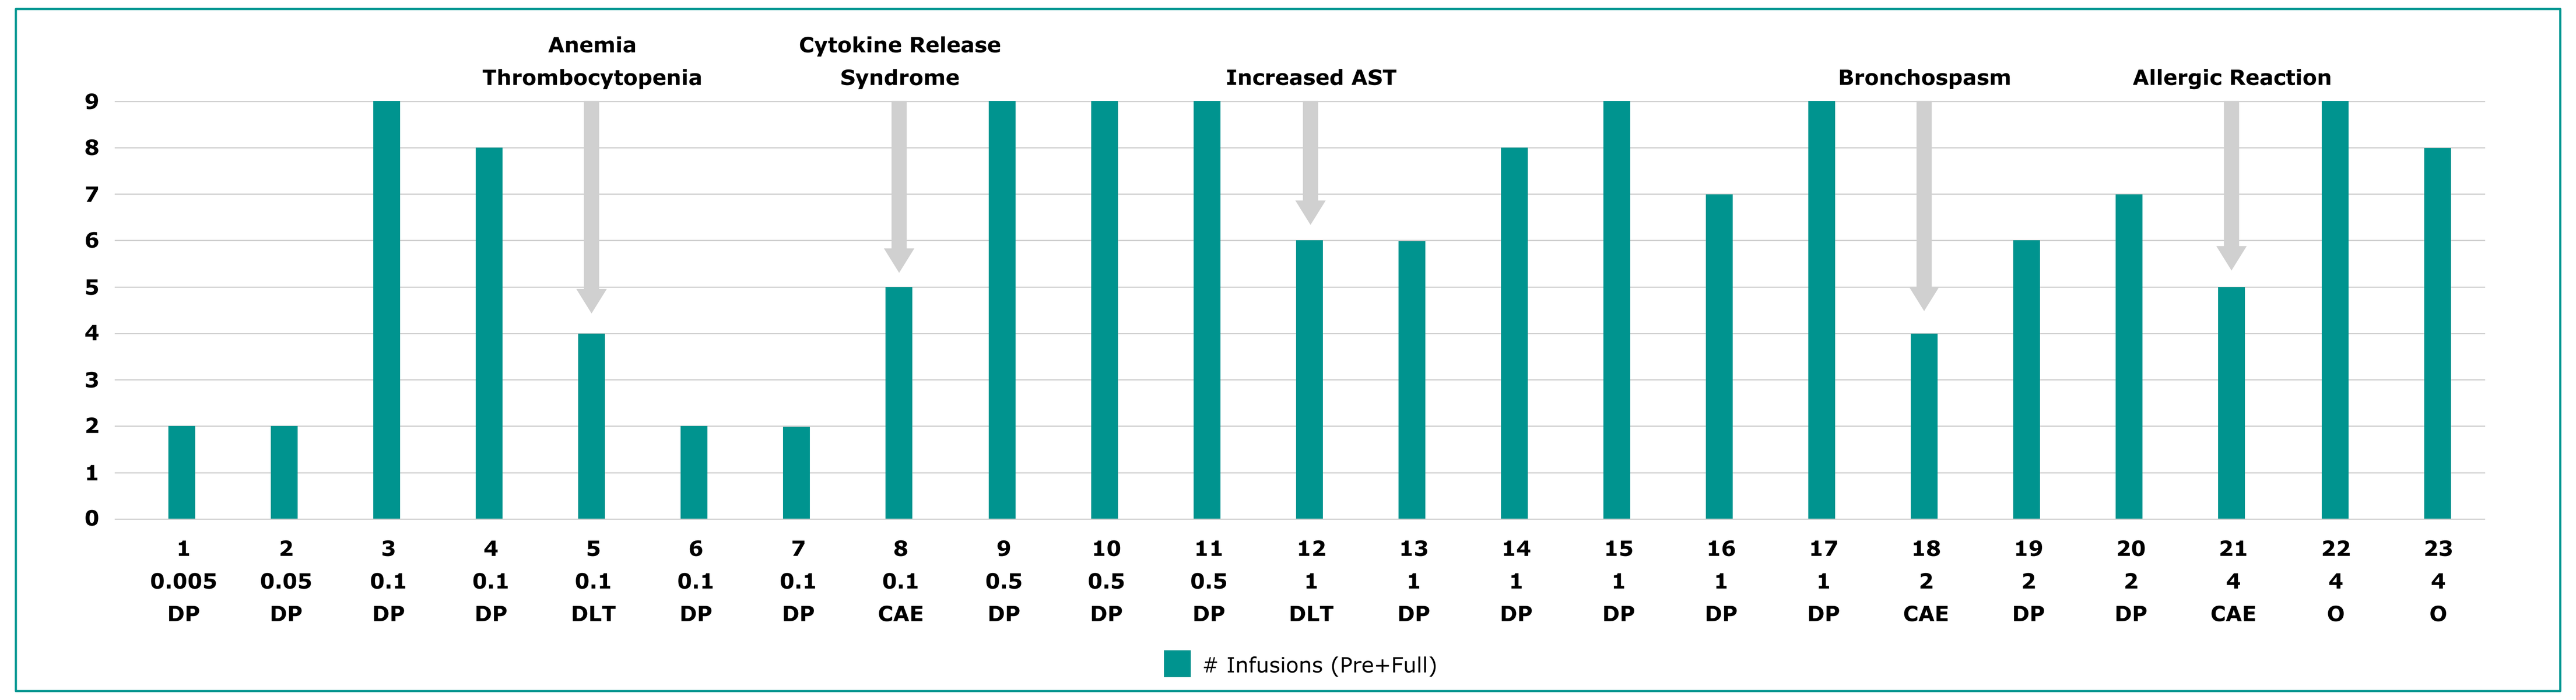
Anemia Thrombocytopenia
Cytokine Release Syndrome
Increased AST
Bronchospasm
Allergic Reaction
0 1 2 3 4 5 6 7 8 9
1 0.005 DP
2 0.05 DP
3 0.1 DP
4 0.1 DP
5 0.1 DLT
6 0.1 DP
7 0.1 DP
8 0.1 CAE
9 0.5 DP
10 0.5 DP
11 0.5 DP
12 1 DLT
13 1 DP
14 1 DP
15 1 DP
16 1 DP
17 1 DP
18 2 CAE
19 2 DP
20 2 DP
21 4 CAE
22 4 O
23 4 O
Infusions (Pre+Full)

Figure 1: The number of daratumumab doses administrated in patients 1 – 23 in the dose-cohorts 0.005 mg/ - 4 mg/kg. DP: disease progression, DLT: dose limiting toxicity, CAE: critical adverse event, O: patient ongoing

Cohort	n/N	Max reduction in M-component in patients with reduction	
		Serum	Urine
≤ 1 mg/kg	3/17	12 % 14 % 19 %	22 % ^ ^
2 mg/kg	1/3	^	55 %
4 mg/kg	3/3	49 % 55 % 61 %*	^ 73 % *^

Table 4: Maximum reduction in M-component. The reduction is measured from baseline until 1 week after last dose. n/N = number of patients with a reduction of M-component/number of patients in the group. *dosing still ongoing. ^ not measureable at baseline.

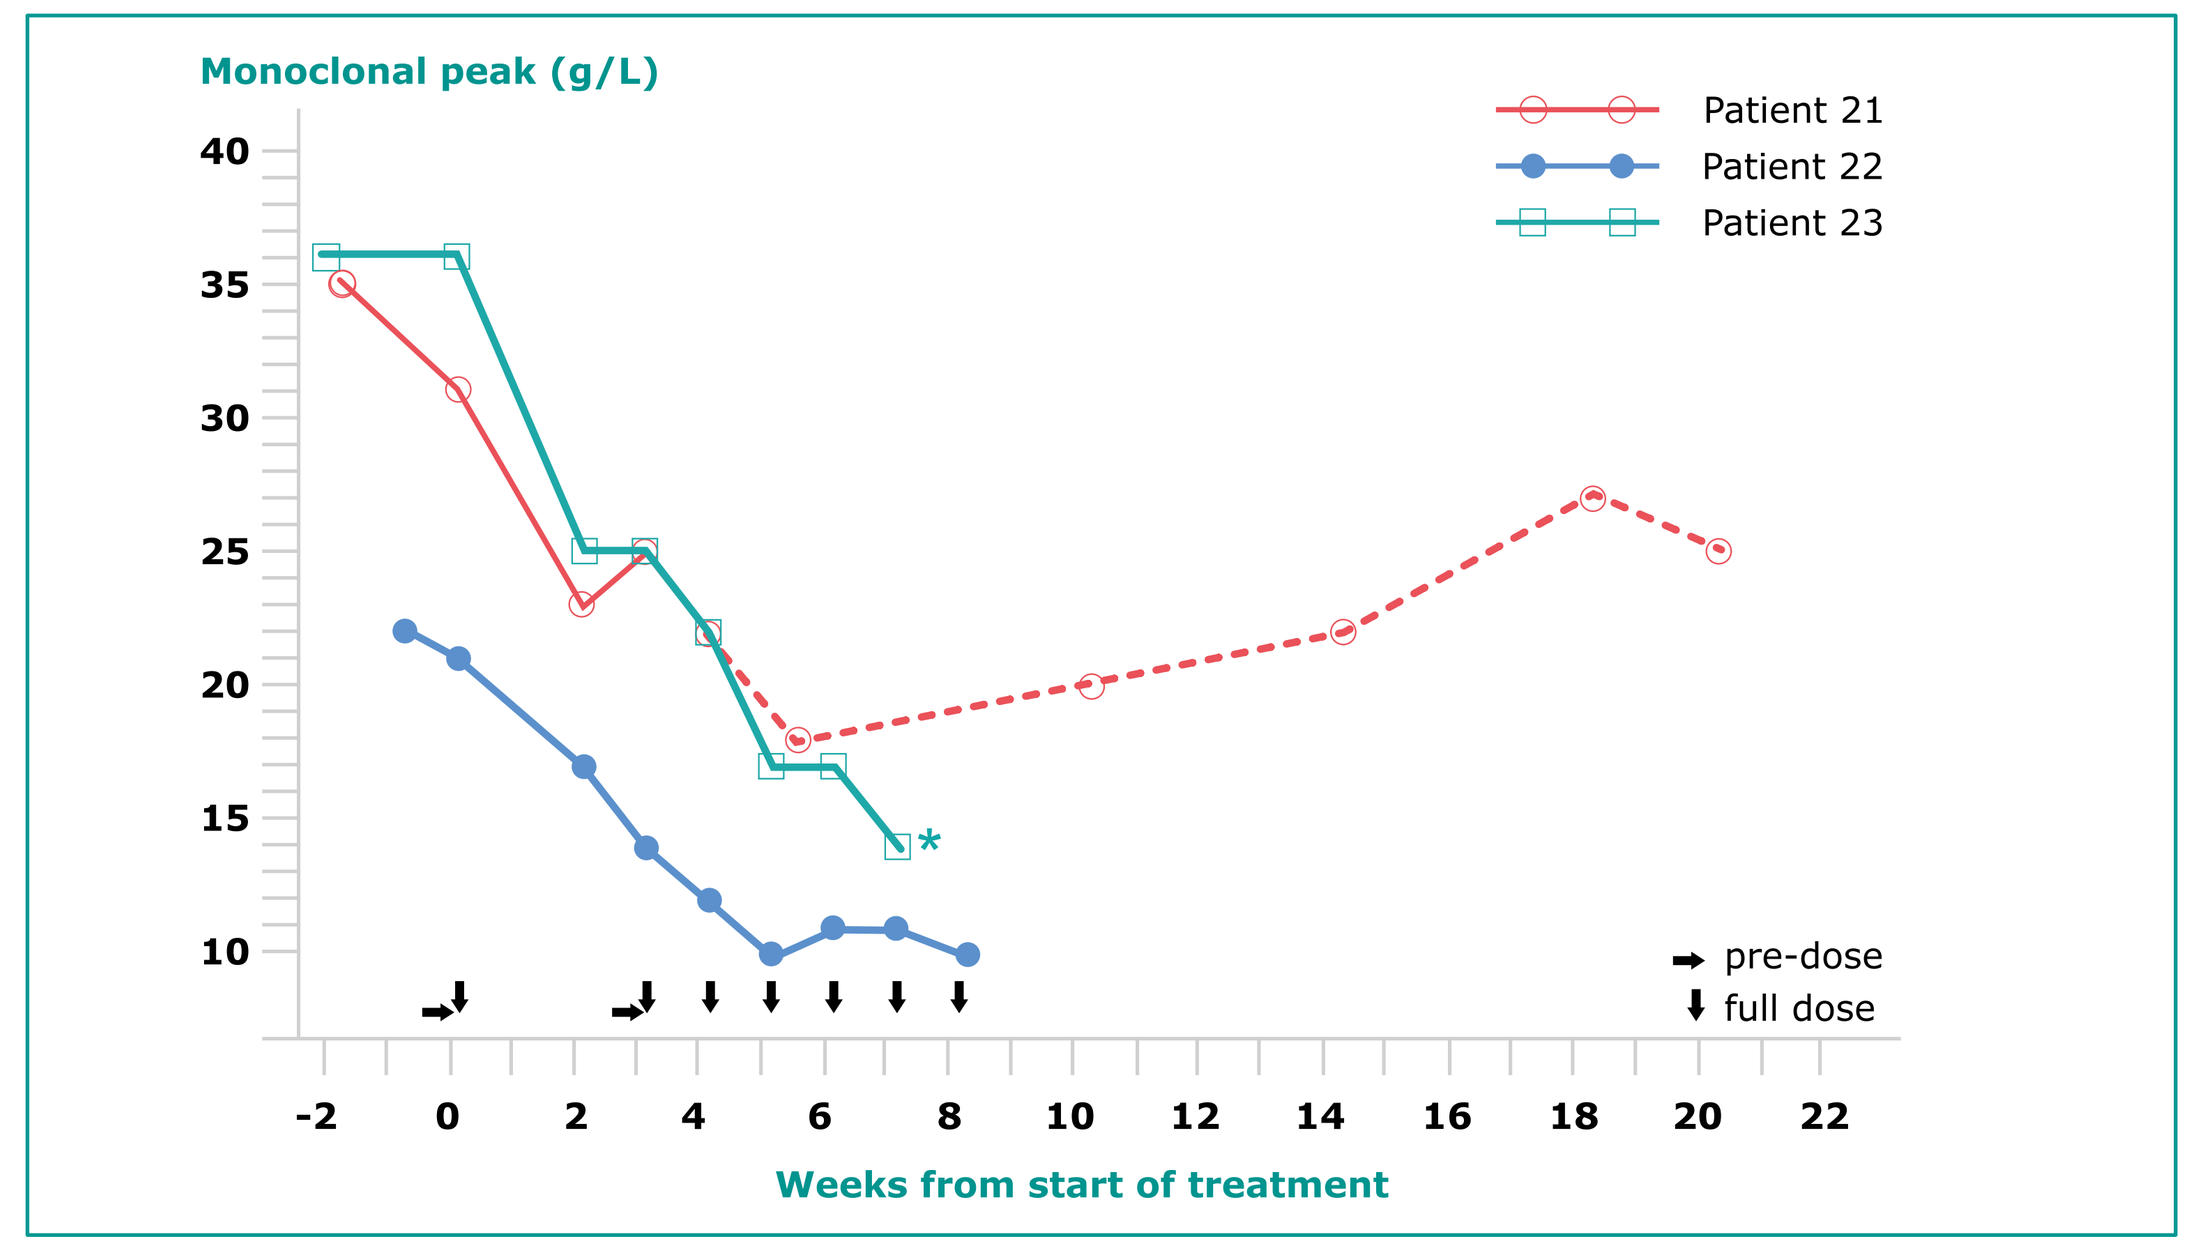
Monoclonal peak (g/L)
40 35 30 25 20 15 10
Patient 21
Patient 22
Patient 23
pre-dose
full dose
-2 0 2 4 6 8 10 12 14 16 18 20 22
Weeks from start of treatment

Figure 2: Reduction in serum M-component during treatment with 4mg/kg daratumumab. Solid line: treatment phase. Dashed line: patient withdrawn, data from follow-up phase, *dosing still ongoing.

CONCLUSION

- Daratumumab has a favorable safety profile as monotherapy in patients with MM in doses up to and including 4 mg/kg.
- The MTD has not yet been reached.
- Based on the observed safety profile, further dose-escalation is ongoing.
- In the 4 mg/kg dose cohort, a 49-61% reduction in the serum M-component is observed.

REFERENCES

1. de Weers et al. J Immunol 2011;186:1840-8.
2. Common Terminology Criteria for Adverse Events v3.0 (CTCAE). Published 2003; December 12th

DISCLOSURES

The study is supported by Genmab A/S, www.genmab.com

Peter Gimsing: There are no relevant conflicts of interest to disclose. Torben Plesner: Genmab A/S: Consultancy. Hareth Nahi: There are no relevant conflicts of interest to disclose. Henk Lokhorst: Genmab A/S: Consultancy. Marie-Louise Valentin and Steen Lisby: Genmab A/S: Employment. Paul Richardson: Genmab A/S: Consultancy.

Abstract #1873 presented at the American Society of Hematology (ASH) 53rd Annual Meeting and Exposition; San Diego, CA, USA; December 10-13, 2011.